CLINICAL EFFICACY OF RADIOCARE® (*Cryptomphalus aspersa*) IN THE PREVENTION AND TREATMENT OF ACUTE RADIODERMATITIS

Santos M, Delgado JM, Rodríguez S, Saez J, Errazquin L, Palacios A, Bouché A, Andreu FJ, Sancho S, de las Heras MH, Alonso A, Escó R, Velasco J, Maciá M, Calvo F, Meirió F. **Gicor**

Grupo Español de Investigación en Oncología Radioterápica.

Introduction

Acute radiodermatitis represent an important skin toxicity factor in patients undergoing radiotherapy, as they cause important loss of life quality for affected patients and cause delays on radiotherapy schedules. These deleterious effects indicate the need of adequate short-term measures to improve the patient's health or at least to block further deterioration.

Up-to-date therapeutic options to prevent or heal adverse effects of radiotherapy are not able to meet the patient's requirements. These include emollients, topic corticosteroids, although these are limited to brief periods of treatment due to their potential adverse effects.

Radiation induces skin damage and reduces its regenerative capability, affecting normal wound healing. Thus, potential treatment should induce, potentiate and/or accelerate the regenerative capability of damaged skin.

The glycoprotein secretion obtained from the mollusc ***Cryptomphalus Aspersa*** **(SCA)**, bears high **Antioxidant Activity** based on its ability to capture free radicals produced during irradiation and also to inhibit their production. In addition, it bears superoxide dismutase as well as gluthation-S-transferase activities.

SCA enhances Proliferation and other Functional Capabilities of Fibroblasts, inducing synthesis of skin elements required for wound healing. SCA increases collagen synthesis, fibronectin deposition on the extracellular matrix and hyaluronic acid content.

Both the facilitating and promoter actions of Radiocare® (SCA) on the mechanisms of cutaneous wound healing provide a rationale for its employment in the treatment of Radiodermatitis

0	I	II	III	IV
No change over baseline	Follicular, faint or dull erythema; epilation; dry desquamation; decreased sweating	Tender or bright erythema, patchy moist desquamation; moderate edema	Confluent, moist desquamation other than skinfolds, pitting edema	Ulceration, haemorrhage, necrosis

Table 1: Skin toxicity scale according to the RTOG (Cutaneous toxicity scale of the Radiation Therapy Oncology Group

Adverse event	I	II	III	IV
Radiation dermatitis	Faint erythema or dry desquamation	Moderate to brisk erythema or a patchy moist desquamation, mostly confined to skin folds and creases; moderate edema	Confluent moist desquamation ≥ 1.5 cm diameter and not confined to skin folds; pitting edema	Skin necrosis or ulcerration of full thickness dermis; may include bleeding not induced by minor trauma or abrasion

Note: Pain associated with radiation dermatitis is graded separately in the PAIN category as Pain to radiation.

Table 2: Skin toxicity scale according to the CTC, version 2.0 (Commo Terminology Criteria for Adverse Events)

Objetive

To evaluate the efficiency and tolerance of Radiocare® in the prevention and treatment of acute radiodermatitis (Grade: I-II) in patients diagnosed with breast carcinoma, about to initiate or undergoing radiotherapy treatment.

Material & Methods

Type of study: Open, controlled, multicentric study.

Population: 96 women, diagnosed with breast carcinoma, mean age of 55 years. Irradiation model: Early breast cancer: (50Gy / 25 fractions of 200cGy / 5wks + 10-16 Gy to the tumor area).

Design: Patients were divided into three arms:

1. **Control arm (Standard treatment): 22 patients**, who received normal treatment, according to the degree of skin toxicity.
 - No toxicity: No treatment.
 - Grade I toxicity: Camomile water
 - Grade II toxicity: Topic corticosteroids (Fluocinolone acetonide) 1/d
2. **Preventive Radiocare® arm: 33 patients**, who received Radiocare® preventively, once a day, from the beginning of radiotherapy to its end.
3. **Curative Radiocare® arm: 41 patients** undergoing radiotherapy, who received Radiocare® if they presented acute radiodermatitis lesions, grades I and II. Every patient within this group received daily treatment with Radiocare from the moment of detection of the wounds until their remission.

Clinical and photographic evaluation:

Initial evaluation, followed by weekly revision during radiotherapy treatment. After completion of the radiotherapy, repeated evaluations were conducted after 1, 2 and 4 weeks. Toxicity was scored according to RTOG and CTC v2.0 criteria (see Tables I and II) and to their symptoms: itching, pain, erythema, desquamation. Score was as follows: 0, absent; 1, mild; 2, moderate; 3, severe.

Evaluation of the level of satisfaction of both the patient and the investigator is referred to different aspects such as product presentation, handling, tolerance and therapeutic efficiency, grouping them in values of a Likert scale from 0 (very good) to 4 (very poor).

Statistics:

Results have been analyzed employing descriptive statistics for every variable included in the study.

For comparison of category variables, Chi-squared or Fisher test was employed, whereas Student's t test or variance (or its non-parametric equivalent) was used. Time-to-event variables were compared with the log-rank test. For multiple comparisons, Tukey or Bonferroni test were applied. Statistics were performed with a 5% significance level.

Results

- **Irradiation dose:** Patients on the curative Radiocare® arm received a mean radiation dose of 32 Gy. 80% patients show GI and 20% show GII.
- **Interruptions of treatment in the three groups:** no significant differences were found in the occurrence (p= 0.330) or in the length (p=0.916) of the interruptions.
- **General evolution of the degree of radiodermatitis in the three arms of the study (Figures 1-2):** Median time of appearance of radiodermatitis is similar for preventive Radiocare® and Standard arms. No significant differences were found in the evolution of the grade of radiodermatitis, since both arms reach GI in the 4th week (30-40 Gy). From this week onwards, patients of the standard arm evolved towards GII more acutely and severely than patients of the preventive Radiocare® arm until the 6th week, when radiotherapy was interrupted and both groups started to ameliorate. However, evolution post-radiotherapy shows a faster and clear-cut tendency towards normalization of the preventive Radiocare® arm compared to the standard group (p<0.089). Such difference is statistically significant according to a contingency table of the evolution from week 1 to 2 post-treatment, which shows that 73.3% of the Radiocare® preventive group evolve from GI to G0 radiodermatitis grade (p<0.05). **These data suggest that Radiocare® preventive treatment allows a faster amelioration of the wounds after radiotherapy**. Curative Radiocare® as well as Standard groups show similar evolution, with a slight, non-significant advantage of the Curative Radiocare® group. Post-treatment evolution was similar for the Curative Radiocare® and Preventive Radiocare®. Evolution has been similar according to RTOG and CTC (Figures 1 and 2).

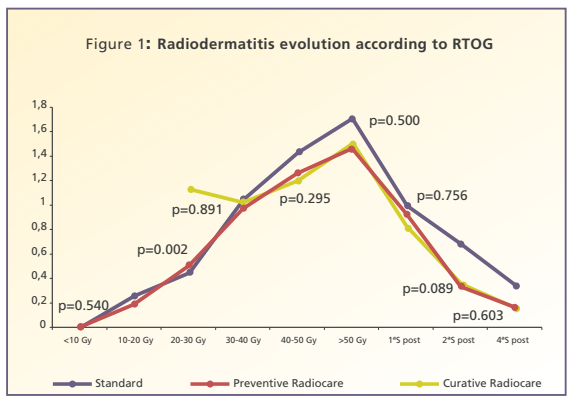
Figure 1: Radiodermatitis evolution according to RTOG
p=0.540
p=0.002
p=0.891
p=0.295
p=0.500
p=0.756
p=0.089
p=0.603
<10 Gy 10-20 Gy 20-30 Gy 30-40 Gy 40-50 Gy >50 Gy 1ªS post 2ªS post 4ªS post
Standard
Preventive Radiocare
Curative Radiocare

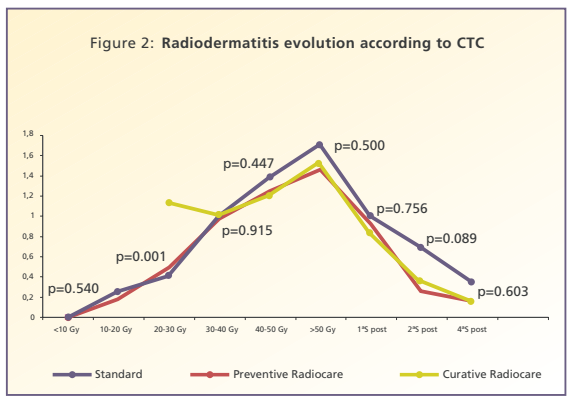
Figure 2: Radiodermatitis evolution according to CTC
p=0.540
p=0.001
p=0.447
p=0.915
p=0.500
p=0.756
p=0.089
p=0.603
<10 Gy 10-20 Gy 20-30 Gy 30-40 Gy 40-50 Gy >50 Gy 1ªS post 2ªS post 4ªS post
Standard
Preventive Radiocare
Curative Radiocare

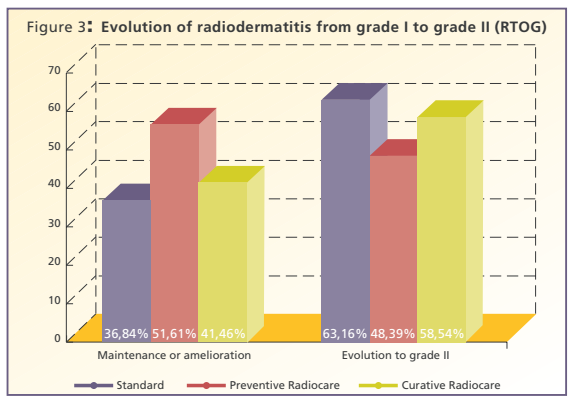
Figure 3: Evolution of radiodermatitis from grade I to grade II (RTOG)
36,84% 51,61% 41,46%
63,16% 48,39% 58,54%
Maintenance or amelioration
Evolution to grade II
Standard
Preventive Radiocare
Curative Radiocare

Figure 3 show that **63.16% of the Standard group** evolves to grade II radiodermatitis, whereas only **48.39% of the preventive Radiocare® group** shows similar behaviour. This is not statistically significant due to the size of the sample, but difference of percentage highlights the beneficial effect of Radiocare® since the beginning of radiotherapy. It is of note that patients of the standard group required corticosteroids upon appearance of GII radiodermatitis. Finally, patients of the Curative Radiocare® group showed slightly lower levels of evolution to GII radiodermatitis compared to the standard group (58.54%).

- **Symptoms and associated signs**

 Evaluation of each one of them has shown similar distribution for all the arms of the study: **Itching,** (p=0,325); **Pain,** (p=0,440); **Erythema** (p=0.911); **Desquamation,** (p=0.138).

- **Evaluation of the level of satisfaction of patients**

 Results are similar up to 40-50 Gy, from there on satisfaction is higher with Preventive Radiocare® than with Curative Radiocare® (p=0.0492). **In summary, patients from both arms with Radiocare® have evaluated treatment better than patients undergoing standard treatment (Figure 4)**.

 According to the worst evaluation made by each individual patient, it can be concluded that **55%** patients of the standard group considered therapy as Very good or Good, compared to **69%** of the Preventive Radiocare® group and **71%** of the Curative Radiocare® group.

- **Evaluation of the degree of satisfaction of the investigator**

 Global evaluation of both treatments with Radiocare® by the investigator has demonstrated to be significantly satisfying (p=0.001). The arms which employed Radiocare® showed similar statistical significance (p=0.8125).

- **Evaluation of tolerance**

 No significant differences were found among the adverse effects appearing in the three groups, and most of them were unrelated to the assigned treatment.

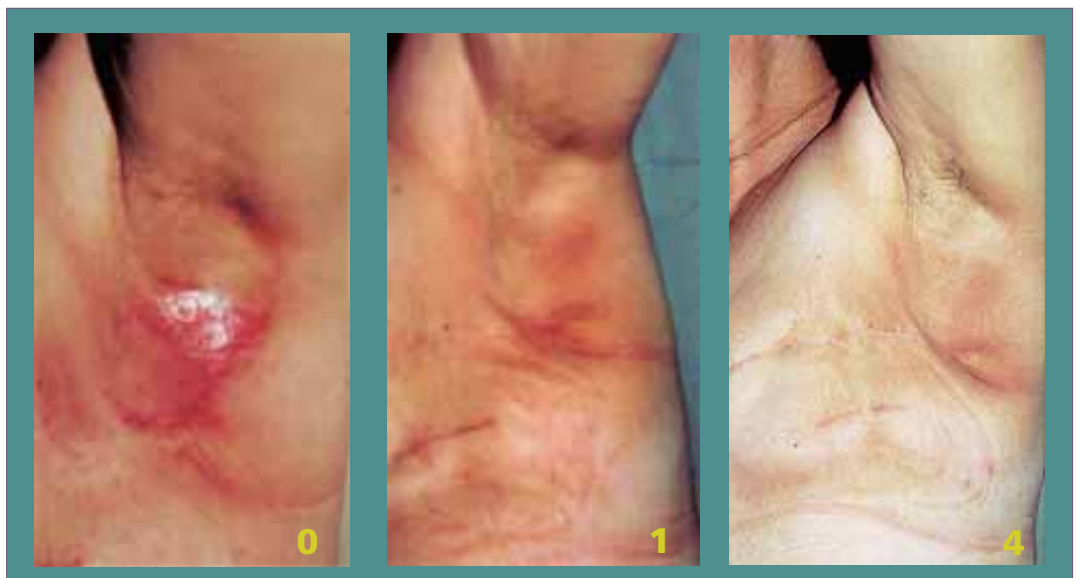
0
1
4

Weeks of treatment with Radiocare

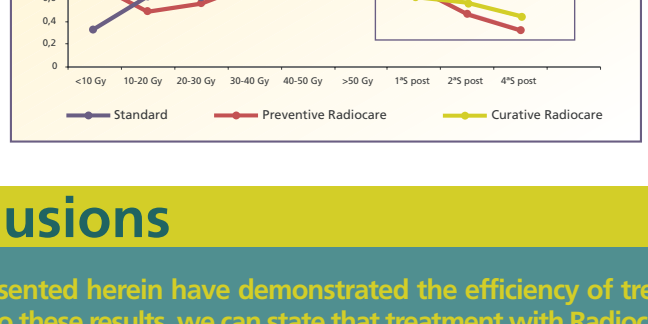
Figure 4: Patient treatment evaluation
p=0.0521
p=0.0428
p=0.0526
<10 Gy 10-20 Gy 20-30 Gy 30-40 Gy 40-50 Gy >50 Gy 1ªS post 2ªS post 4ªS post
Standard
Preventive Radiocare
Curative Radiocare

Comparison of treatments including Radiocare® compared to standard was statistically significant towards Radiocare® (p=0.0001)

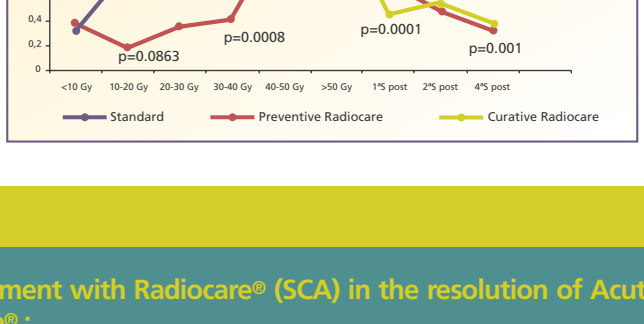
Figure 5: Investigator evaluation
p=0.0863
p=0.0008
p=0.0716
p=0.0001
p=0.001
<10 Gy 10-20 Gy 20-30 Gy 30-40 Gy 40-50 Gy >50 Gy 1ªS post 2ªS post 4ªS post
Standard
Preventive Radiocare
Curative Radiocare

Conclusions

Results presented herein have demonstrated the efficiency of treatment with Radiocare® (SCA) in the resolution of Acute Radiodermatitis, in a statistically similar fashion or even better compared to standard treatment. According to these results, we can state that treatment with Radiocare® :

- **Does not cause adverse effects, not even during prolonged therapy.**
- **It does not interfere with radiotherapy.**
- **Employed in a Curative or Preventive way, it is as efficient than Standard treatment (camomile water and corticosteroids) or even better.**
- **Preventive treatment is significantly more beneficial regarding evolution of radiodermatitis.**
- **Radiocare has been favoured by both Patients and Investigators, being more satisfactory than Standard treatment.**

The regenerative mechanism of action of SCA results in clinical amelioration of lesions caused by Acute Radiodermatitis, supporting its predominance over emolients.
The lack of local and/or sistemic adverse effects in long treatments provide a rationale for election of Radiocare instead of topic corticosteroids.

References

1) Wells M., MacBride S. Radiation skin reactions In Supportive care in radiotherapy. Faithfull S, Wells M. Chapter 8: 135-159. Elseiver Science Limited. 2003 2) Brieva A, Guerrero A, Pivel JP. Un adaptógeno natural para la piel. Dermatol. Cosmet. 1998.1-7 3) Brieva A, Guerrero A, Pivel JP. Mecanismos bioquímicos y farmacológicos relacionados con la actividad de la secreción de Cryptomphalus Aspersa (SCA) en radiodermitis. Dermatol.Cosmet.2001;11(2):71-75 4) Falabela AF, Falanga V. Wound Healing in Freinkel RK and Woodley DT.The Biology of the skin. Chapter 19: 281-297. The Parthenon Publishing Group.2001 5) Efecto de la secreción de Cryptomphalus Aspersa en el ensamblaje de la matriz exracelular de fibronectina. Data on file IFC.2001 6) Schmuth MS; Wimmer MA, Hofer S, Sztankay A, Weinlich G, Linder DM, Elias PM, Fritsch PO, Fritsch E. Topical corticosteroids therapy for acute radiation dermatitis: a prospective, randomised, double - blind study. Br J Dermatol 2002;146:983-991 7) Ledo E, de las Heras ME, Ledo A. Treatment of acute radiodermitis with Cryptomphalus Aspersa Secretion. Radioprotección; 1999; 23 (VII)